Power calculations for time-to-event outcomes using simulation in the presence of improved standard of care in the COVID-19 pandemic.

J.R.J. Inshaw[1], A. Nickless[1], N. Madeleine[2], S. McPherson[2], B. Ferrari[2], A. Jackson[2], S.R. Miller[1]

1: Exploristics Ltd, Belfast, UK
2: BerGenBio, Bergen, Norway

Problem

- Early COVID-19 trials took place prior to the approval of therapies to treat the disease.
- A variety of factors could change the response for the control arm vs. the early trials.

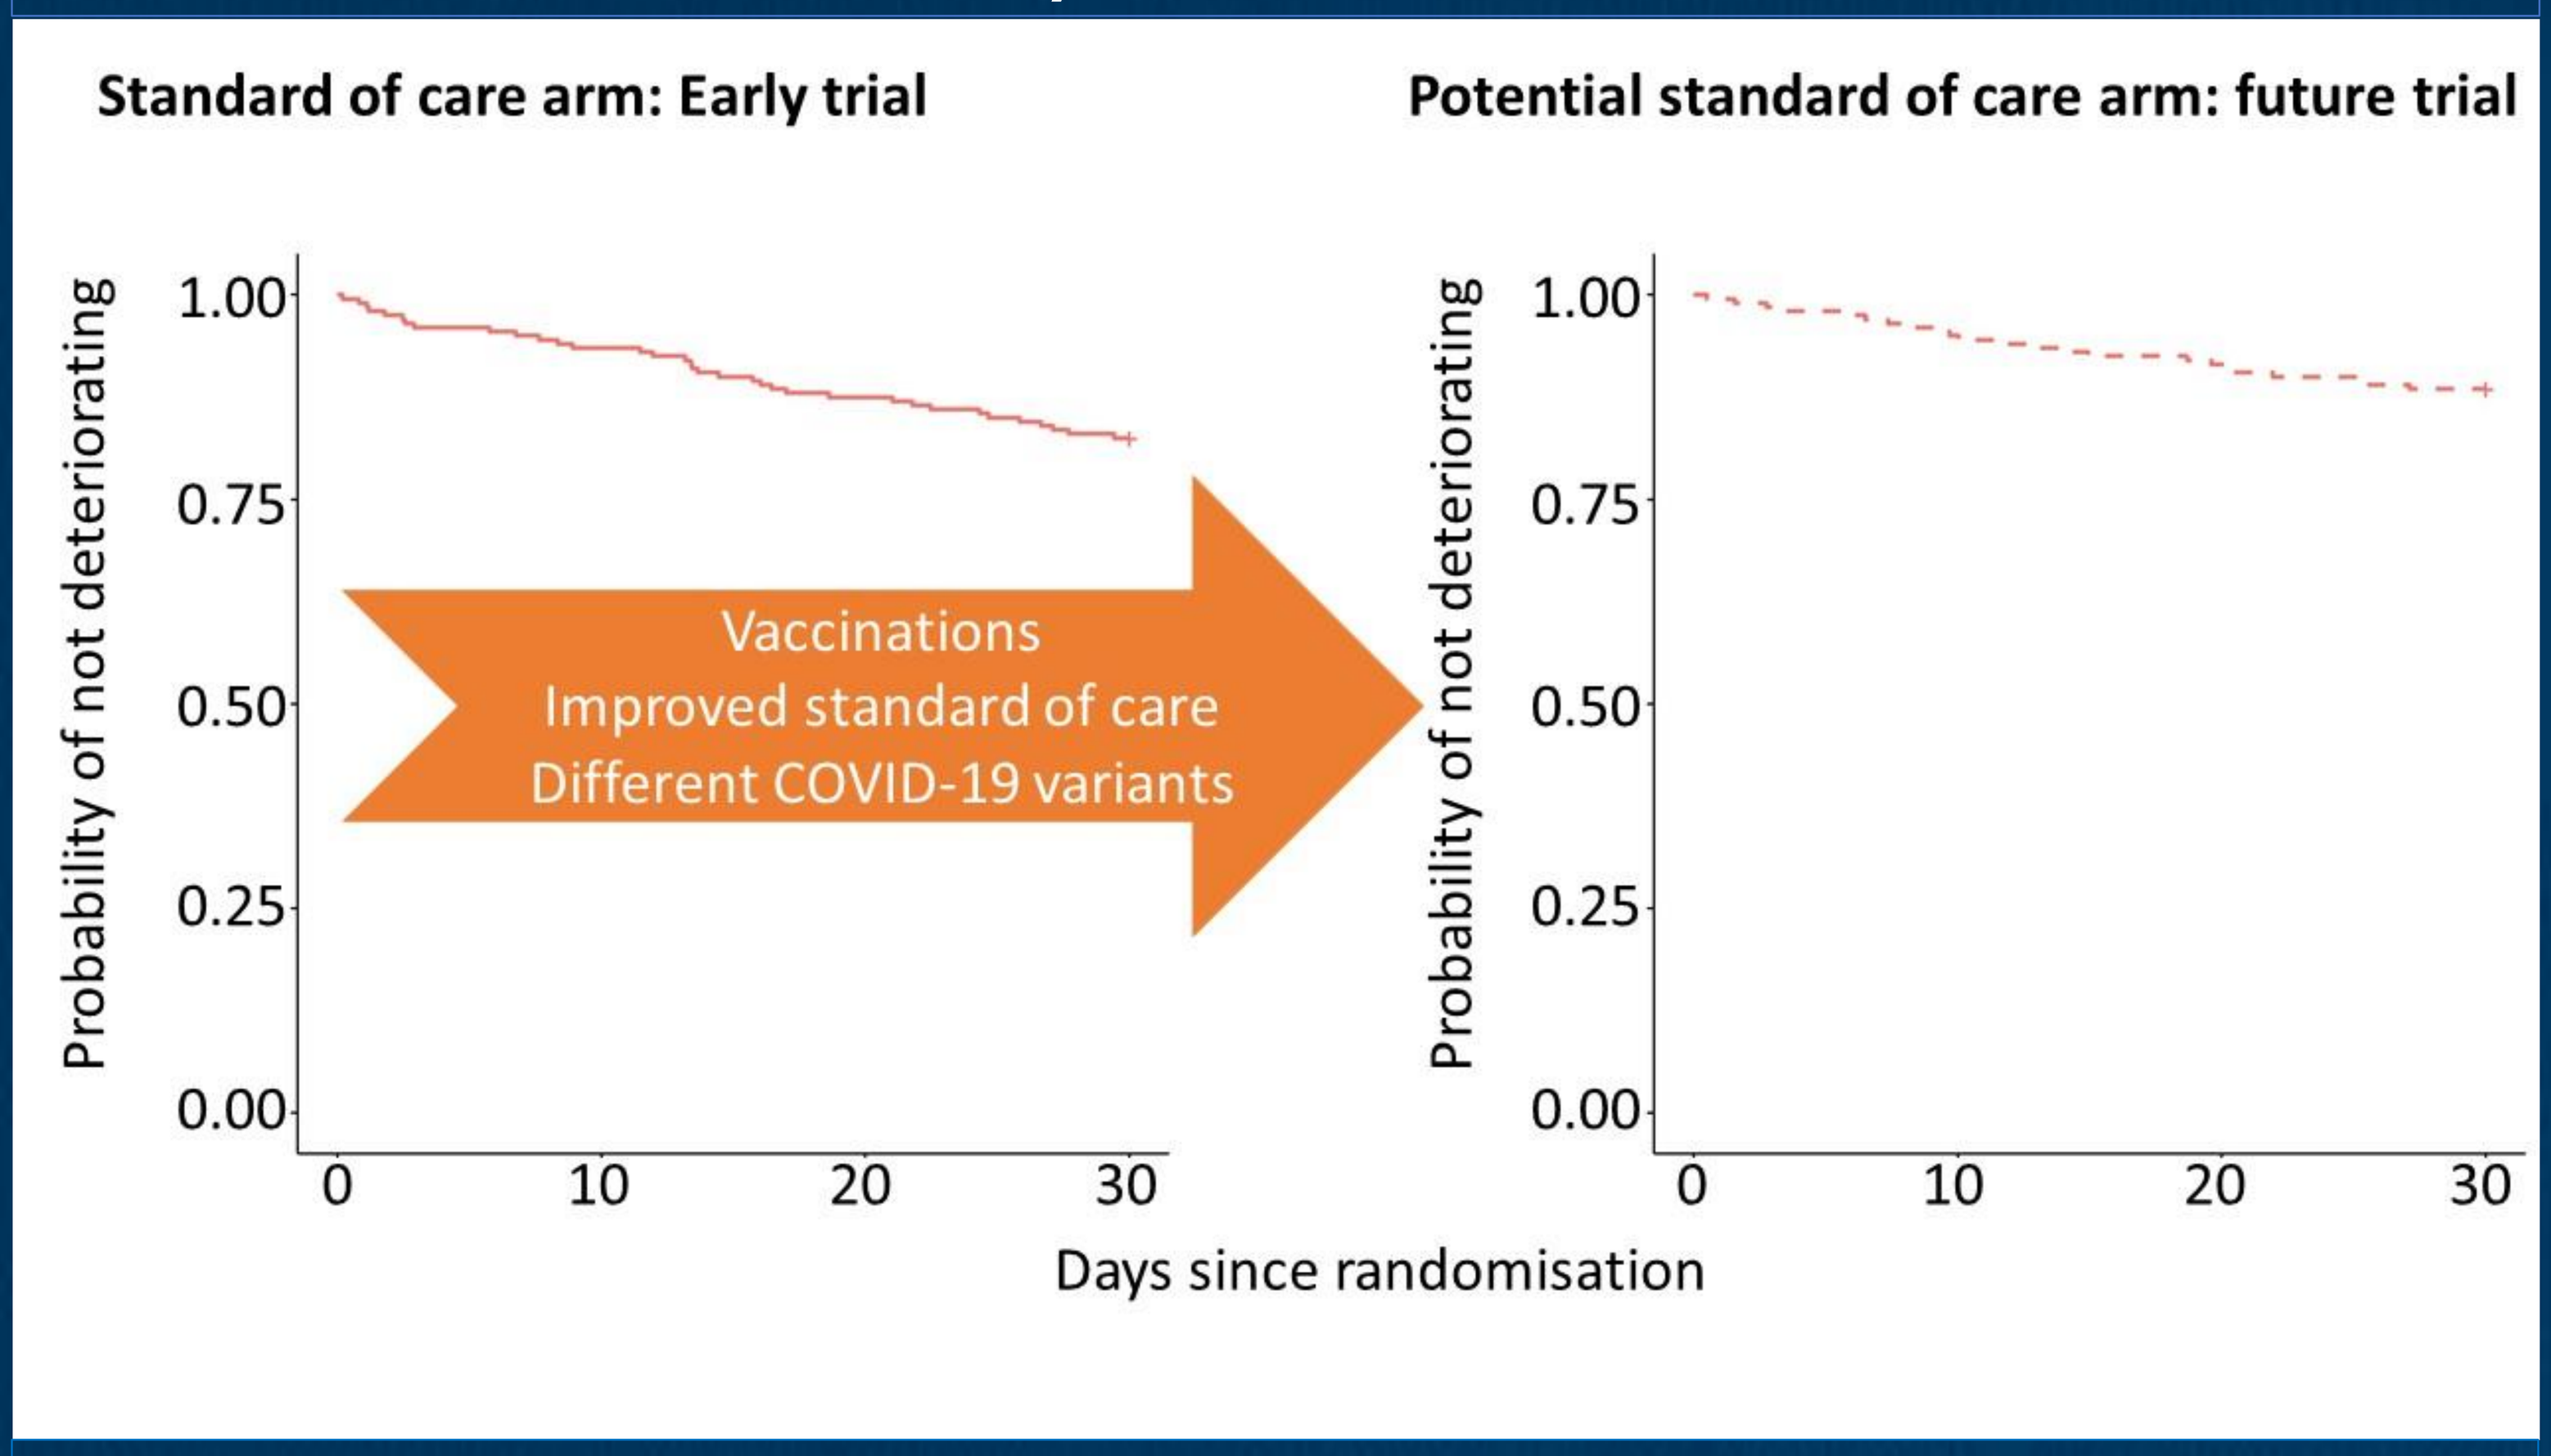
Standard of care arm: Early trial
Potential standard of care arm: future trial
Probability of not deteriorating
1.00
0.75
0.50
0.25
0.00
0
10
20
30
Vaccinations
Improved standard of care
Different COVID-19 variants
Days since randomisation

- Failure to account for this potential change in the control arm may result in low probability of success in future trials.

Aims and approach

- To design a phase IIb study, examining the effect of Bemcentinib on hospitalised COVID-19 patients.
- Endpoint considered was the time to deterioration of 1 point in the WHO ordinal scale over 30 days follow-up.
- We simulated data using *KerusCloud* to mimic a range of possible scenarios. 2000 trials per scenario.
- We held the treatment effect constant at that observed from meta-analysis of two completed phase IIa studies, with a hazard ratio = 0.431, participants less likely to deteriorate on Bemcentinib vs. standard of care.
- Event times for both arms were approximated using Weibull distributions[1], censoring distribution as a uniform at 30 days.
- "Success" defined as log-rank p-value <0.05.

Scenario	Event rate	Description
One	18%	Assuming no change in event rate from early phase trials to future trial.
Two	Patients 0-20%: 18% Patients 20-40%: 16% Patients 40-60%: 14% Patients 60-80%: 12% Patients 80-100%: 10%	Assuming diminishing event rate throughout the course of the future trial.
Three	10%	Assuming lowest event rate throughout the course of the future trial.

Virtual populations

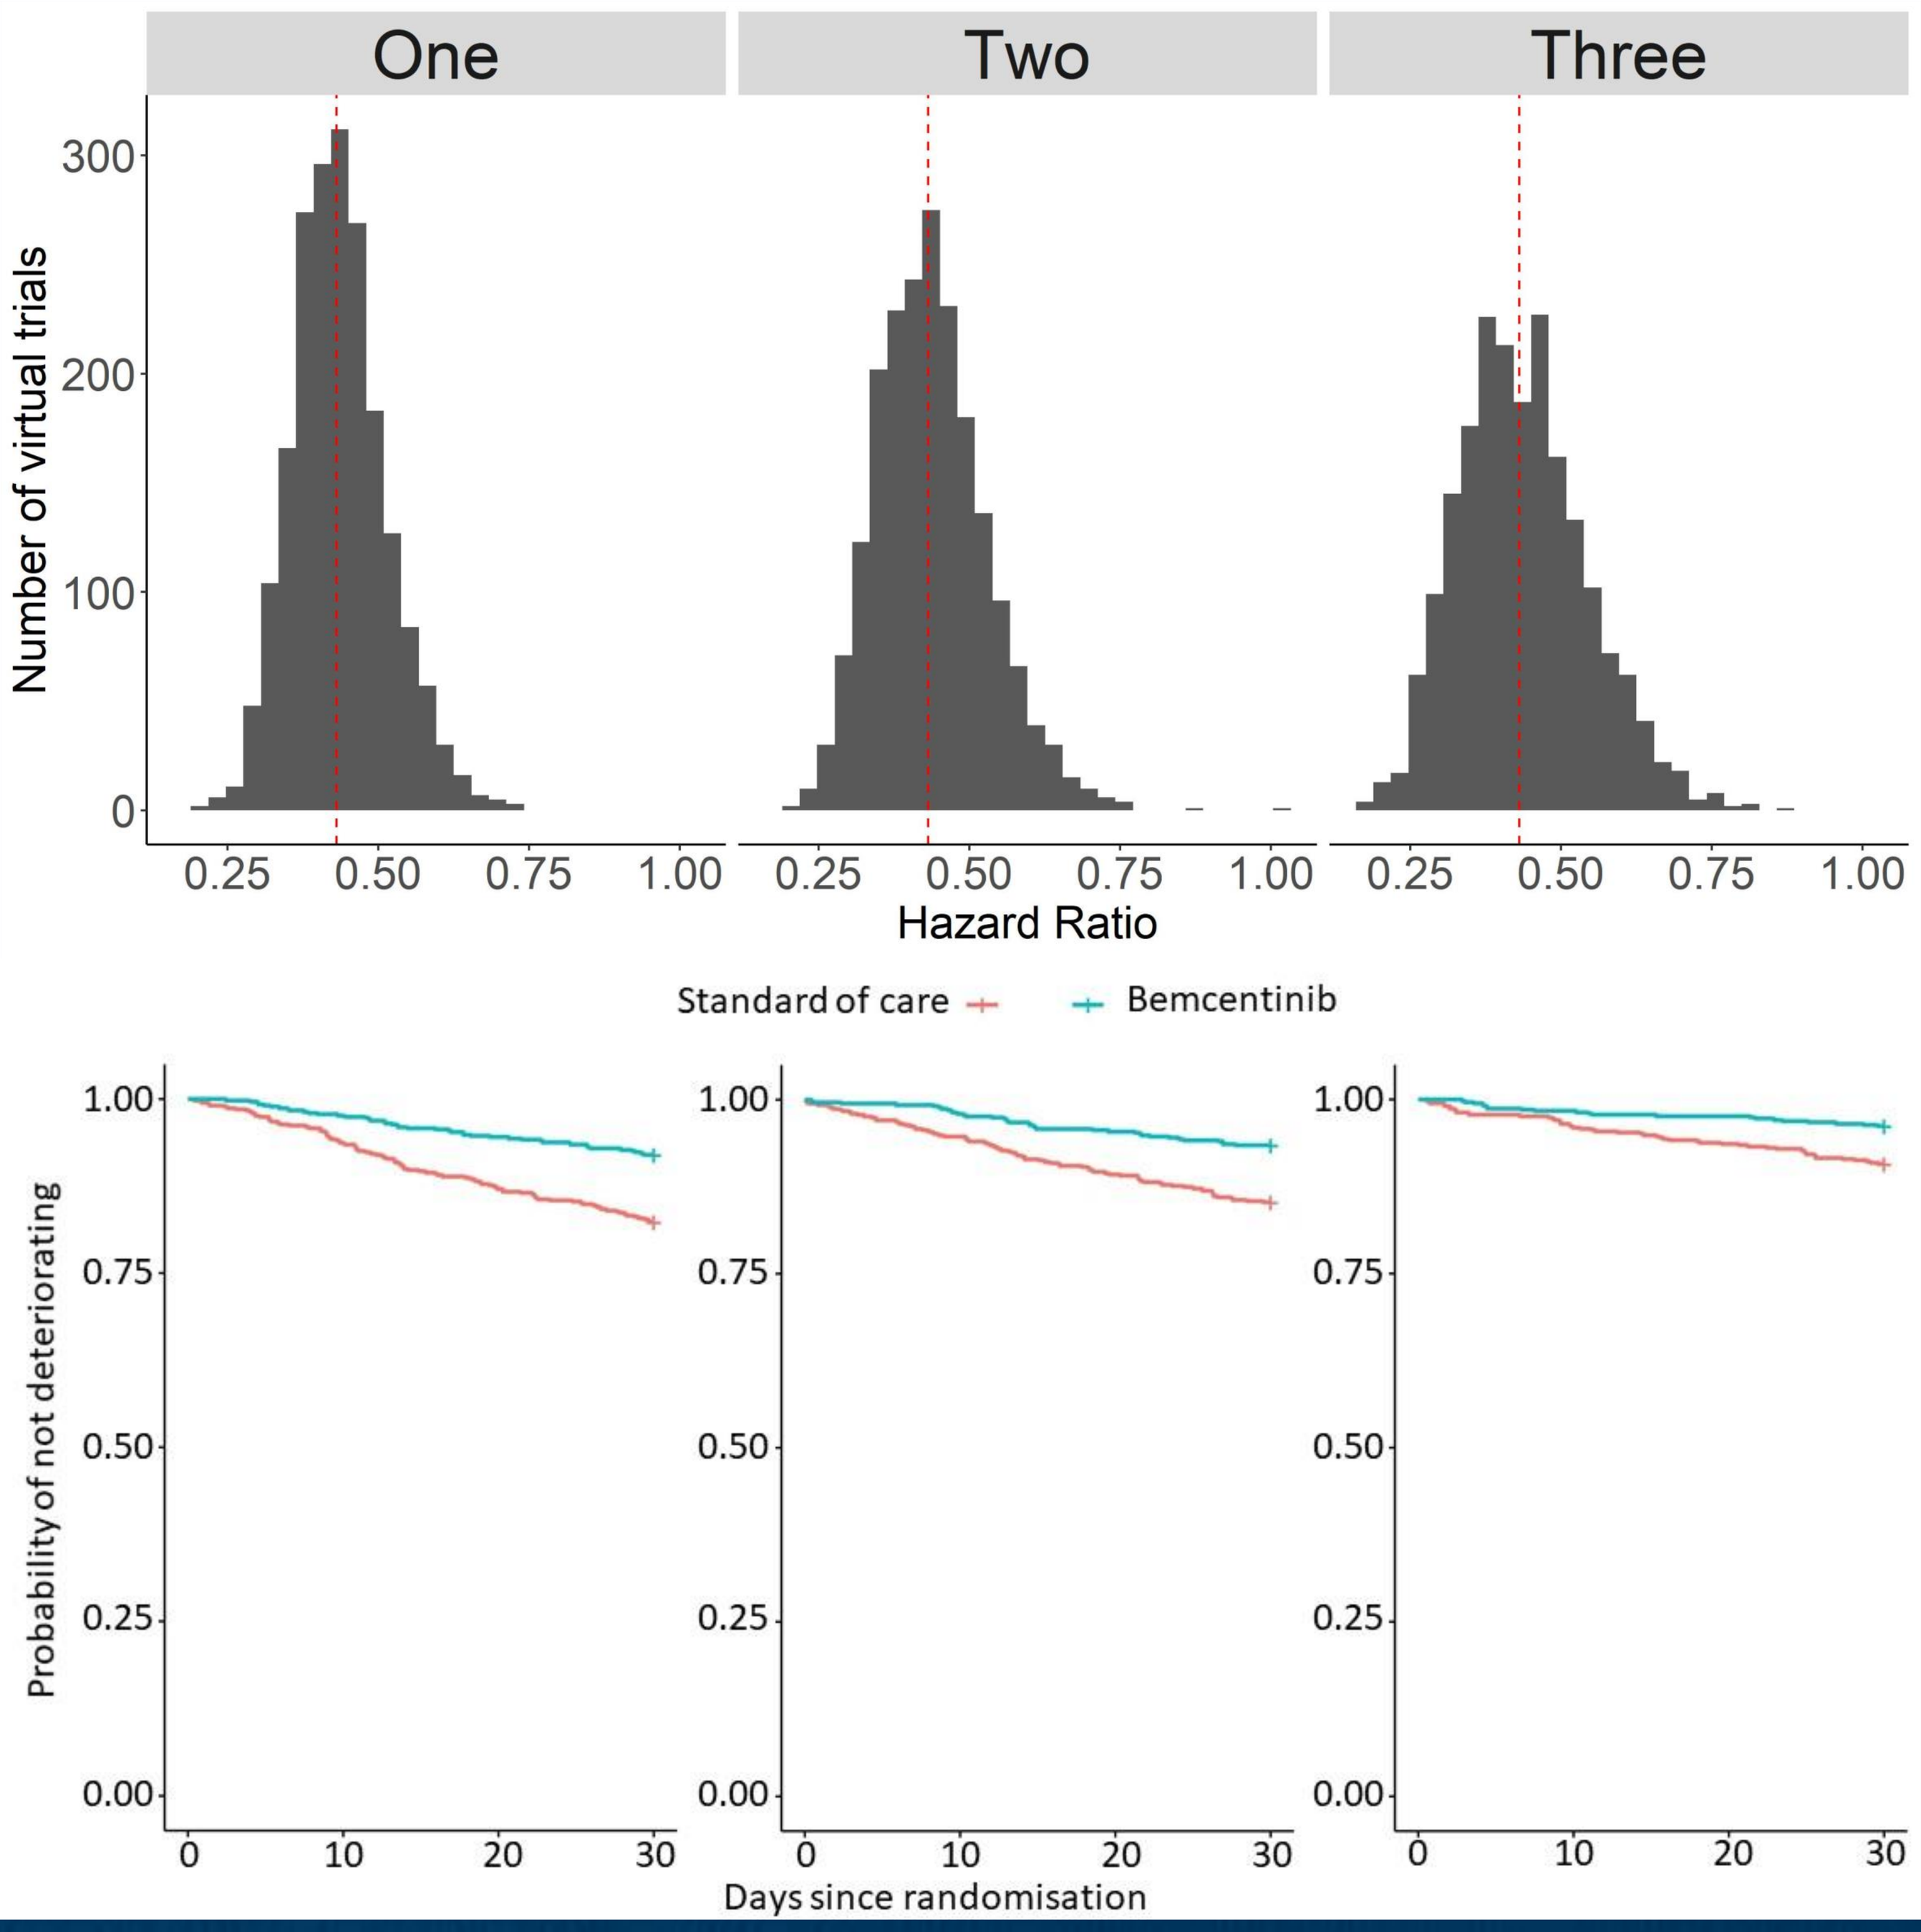
One
Two
Three
Number of virtual trials
300
200
100
0
0.25
0.50
0.75
1.00
Hazard Ratio
Standard of care
Bemcentinib
Probability of not deteriorating
1.00
0.75
0.50
0.25
0.00
0
10
20
30
Days since randomisation

Probabilities of success

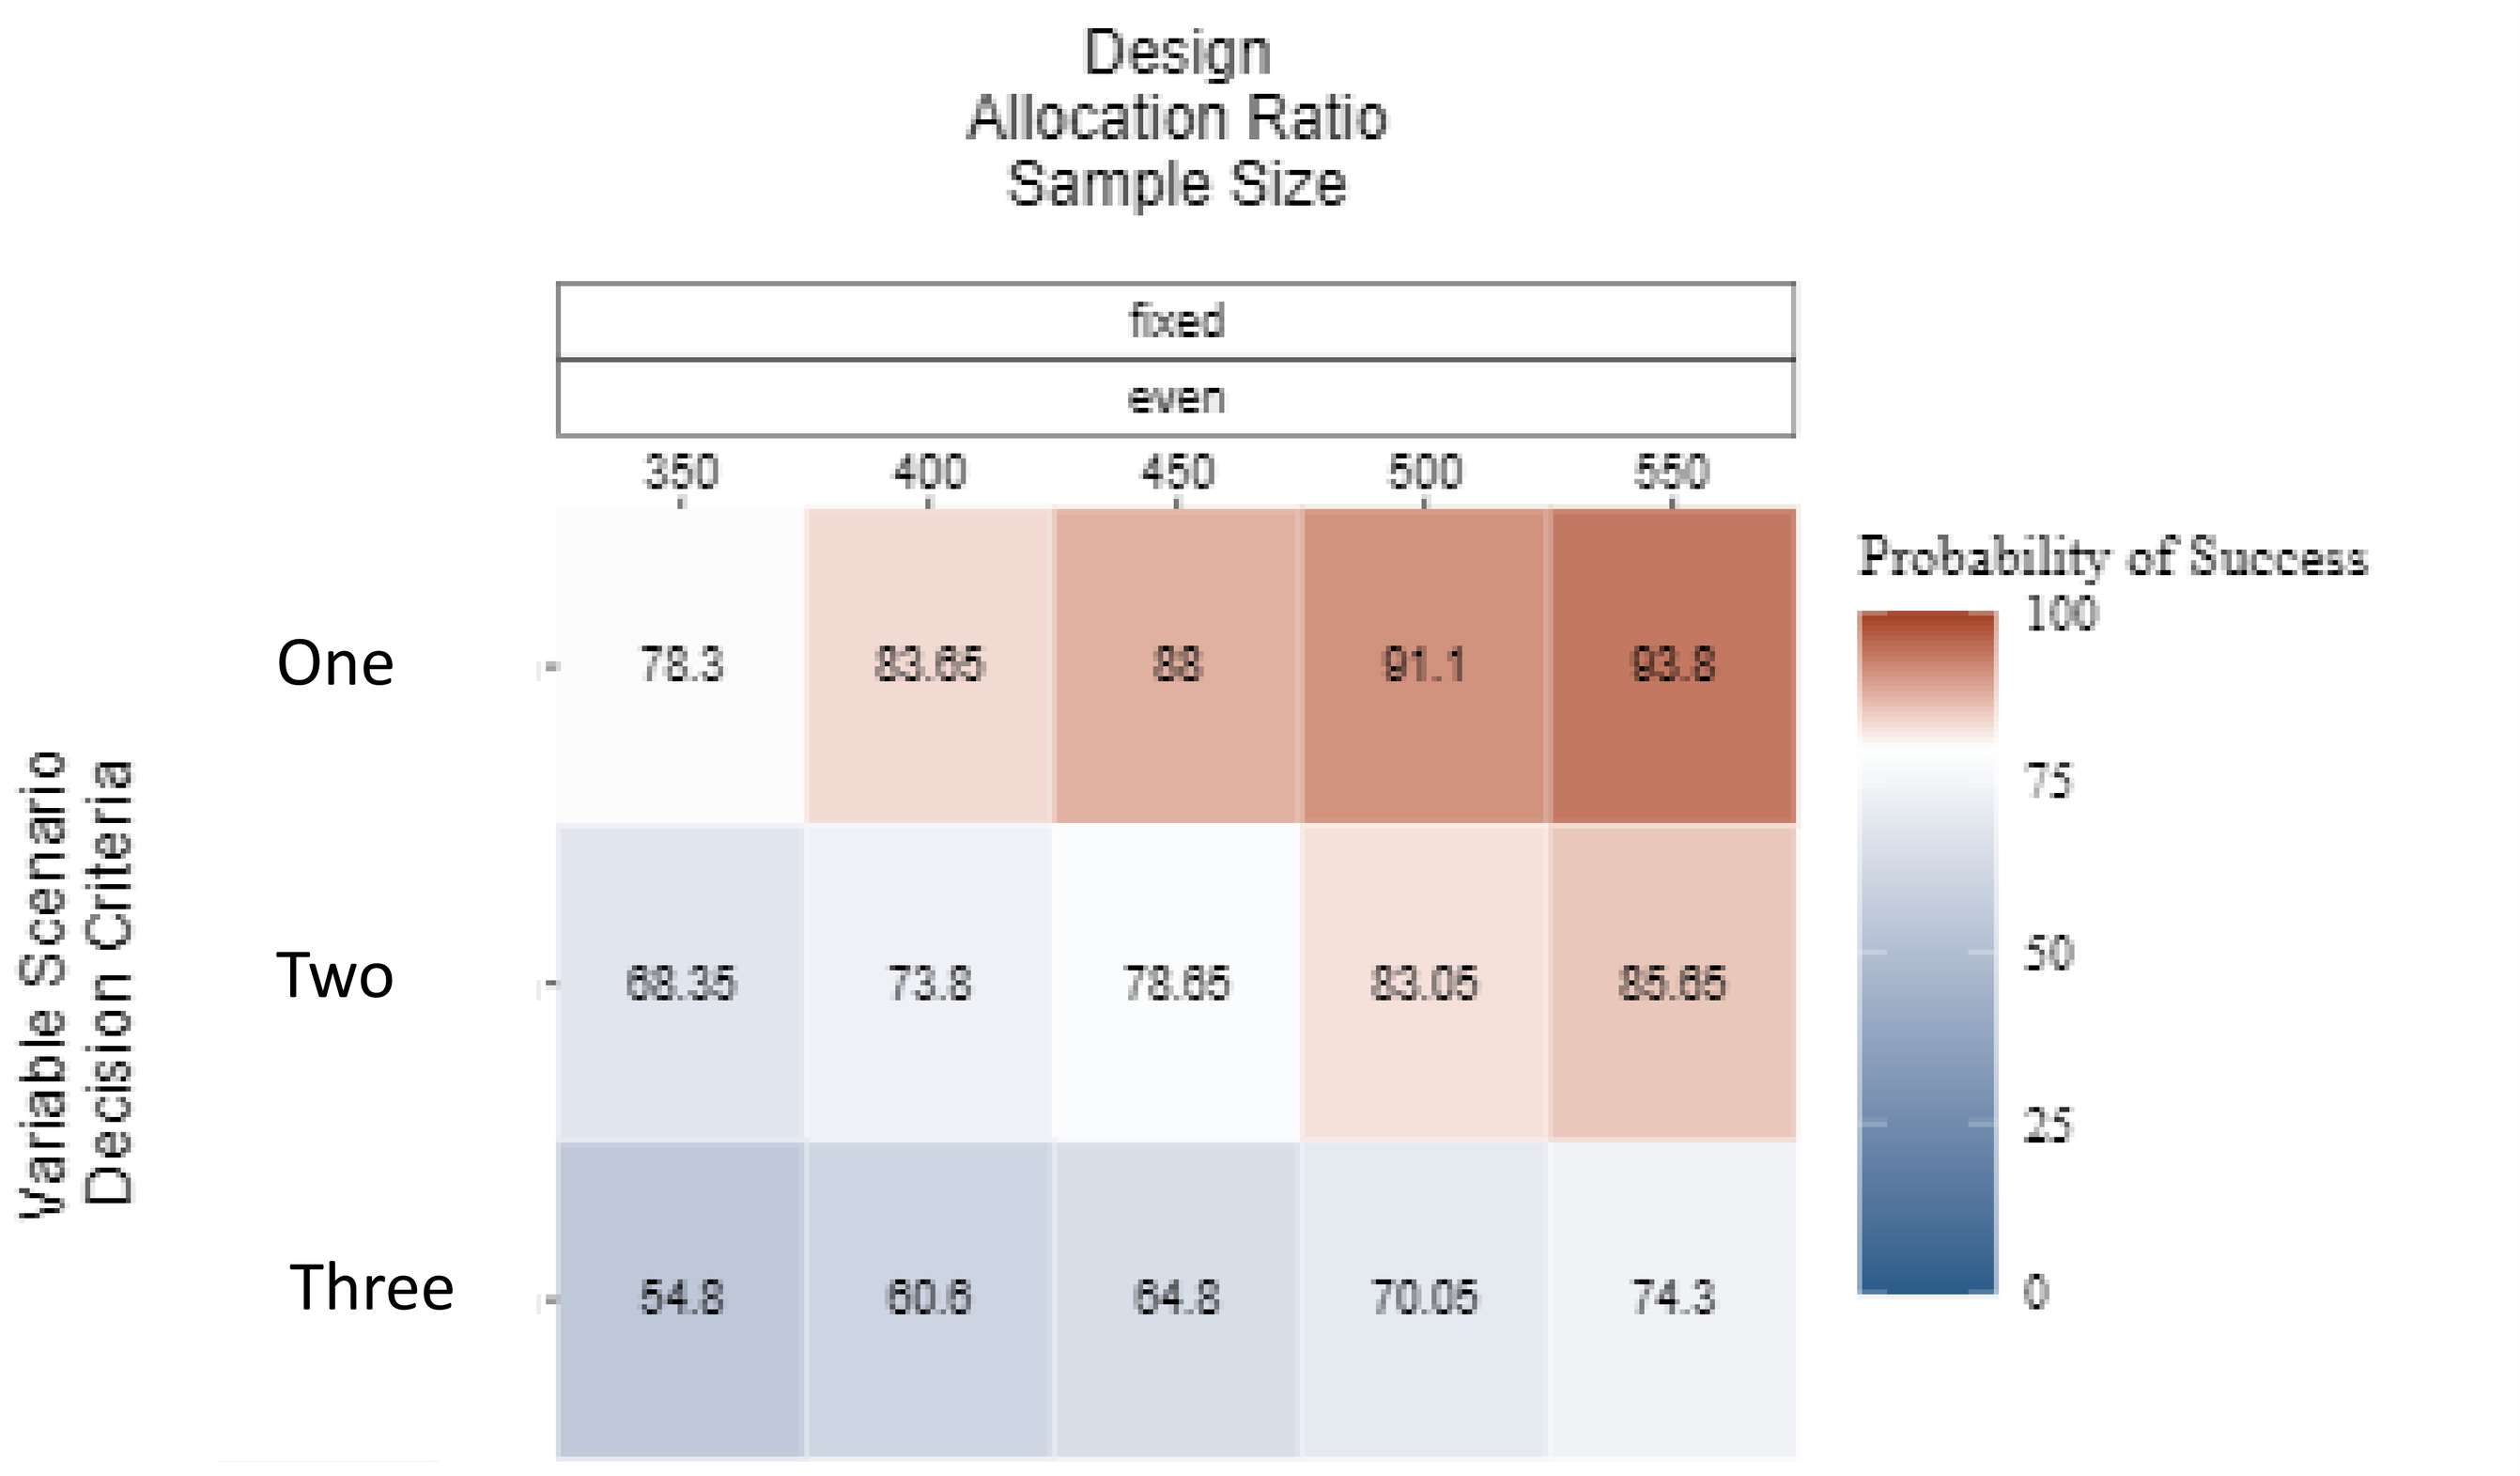
Design
Allocation Ratio
Sample Size
fixed
even
350
400
450
500
550
Variable Scenario
Decision Criteria
One
78.3
83.65
88
91.1
93.8
Two
68.35
73.8
78.65
83.05
85.65
Three
54.8
60.6
64.8
70.05
74.3
Probability of Success
100
75
50
25
0

Conclusions

- Even if the treatment effect size remains consistent between an early trial and the future trial, a change in control arm response can have a large impact on study probability of success.
- If not considered, the probability of success recruiting approximately 350 participants in the future trial would appear to be close to 80%, but if the event rate was notably lower than in the original trial were true (scenario three), it could be closer to 50%.
- Simulation allows quantification of the probability of success under complex scenarios (e.g. scenario two, with diminishing event rate over time) in a way traditional power calculations cannot do.

exploristics

References

[1.] Marks, N.B. Estimation of Weibull parameters from common percentiles. *Journal of applied statistics*, doi: 10.1080/02664760420003051 22

Acknowledgements

- We are grateful to all trial participants.

unlocking the value in data